Nitric Oxide is a Powerful Anti-Coronavirus Inhaled Agent that Acts Within Hours

Matan Goldshtein[2], Omer Lerner[2], Rinat Kalaora[2], Shay Yarkoni[2], Elya Dekel[2], Hila Confino[2], Pam Golden[1], David Greenberg[1,3,4], Steve Lisi[1], Amir Avniel[1,2], Yonat Shemer-Avni[3,4]

[1]Beyond Air® Inc., [2]Beyond Air® Ltd.,[3]Ben Gurion University of the Negev, [4]Soroka Medical Center Contact information: mgoldshtein@beyondair.net

Introduction: Nitric oxide (NO) is a small endogenous messenger molecule with free radical characteristics that plays a key role in the physiological processes in the lung, including host defense against airway pathogens. NO donors have been reported to inhibit replication of severe acute respiratory syndrome coronavirus (SARS-CoV-1) *in vitro*, and exogenous gaseous NO (gNO) at concentrations ≥ 150ppm has been shown to act as a potent antimicrobial agent. In multiple clinical trials, administration of high dose (150 to 250ppm) intermittent gNO (30-40 min cycles, 2-5 cycles a day) was safe and well-tolerated, with promising antimicrobial efficacy potential. Beyond Air® has developed the LungFit™ platform system, which synthesizes up to 400ppm gNO from ambient air and delivers it to the human lung, eliminating the need for cylinders. Given its safety and antimicrobial activity, inhaled gNO is a potential treatment for patients with COVID-19.

Aim: To determine the effects of gNO on human coronavirus infectivity *in vitro* in a proof-of-concept evaluation of the use of gNO as a potential method of preventing and treating human SARS-CoV-2 infection.

Methods: OC43 human coronavirus was exposed *in vitro* to 150-250ppm gNO for up to 8 hours intermittently (1-hour alternating) both before and after infection of human HCT-8 cells. Host cell viability was assessed by an XTT cell proliferation-based assay 3-7 days post exposure to NO. The coronavirus infectivity was assessed by TCID50 (Median Tissue Culture Infectious Dose) 3-7 days post exposure.

Results: When coronavirus was exposed to 250ppm NO for 2 hours prior to infection, a significant reduction in infectivity was achieved as viral load was reduced by 24-fold compared to untreated sample and host cell viability was increased by 75% ($p<0.05$). Post-infection exposure, for 4 hours total, of OC43 coronavirus to 250ppm NO resulted in a 46% increase in host cell viability ($p<0.05$). Upon exposure of coronavirus infected cells to 150ppm gNO, while coronavirus lost 50% of its infectivity after 4 hours of treatment with 150ppm NO, complete inhibition of infectivity was achieved after 8 hours of treatment.

Treatment of host cells infected with coronavirus with 150 - 250 ppm NO improves cell viability and reduces infectivity

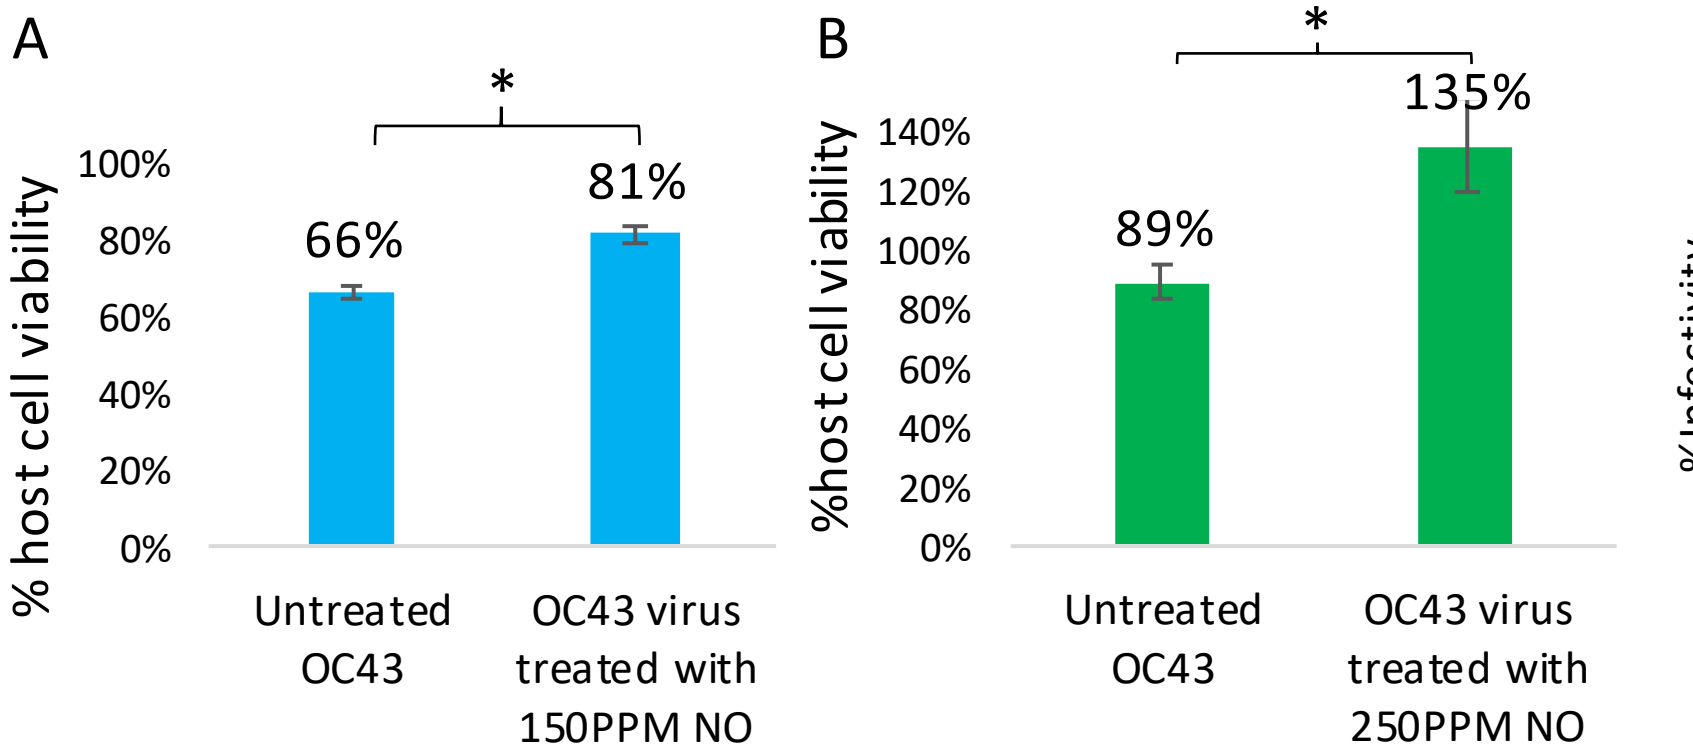
A
% host cell viability
0%
20%
40%
60%
80%
100%
66%
81%
*
Untreated OC43
OC43 virus treated with 150PPM NO
B
%host cell viability
0%
20%
40%
60%
80%
100%
120%
140%
89%
135%
*
Untreated OC43
OC43 virus treated with 250PPM NO

Figure 1: Exposure of OC43 coronavirus infected cells to 150-250 ppm NO improves cell viability. Viability of HCT-8 host cells infected with OC43 human coronavirus 3 days post treatment of 4 cycles of 1-hour exposure to NO and 1 hour in the incubator. A) 150 ppm NO B) 250 ppm NO (n=3, mean+SD, normalized to non-treated cells, *$p<0.05$, Student's t-test)

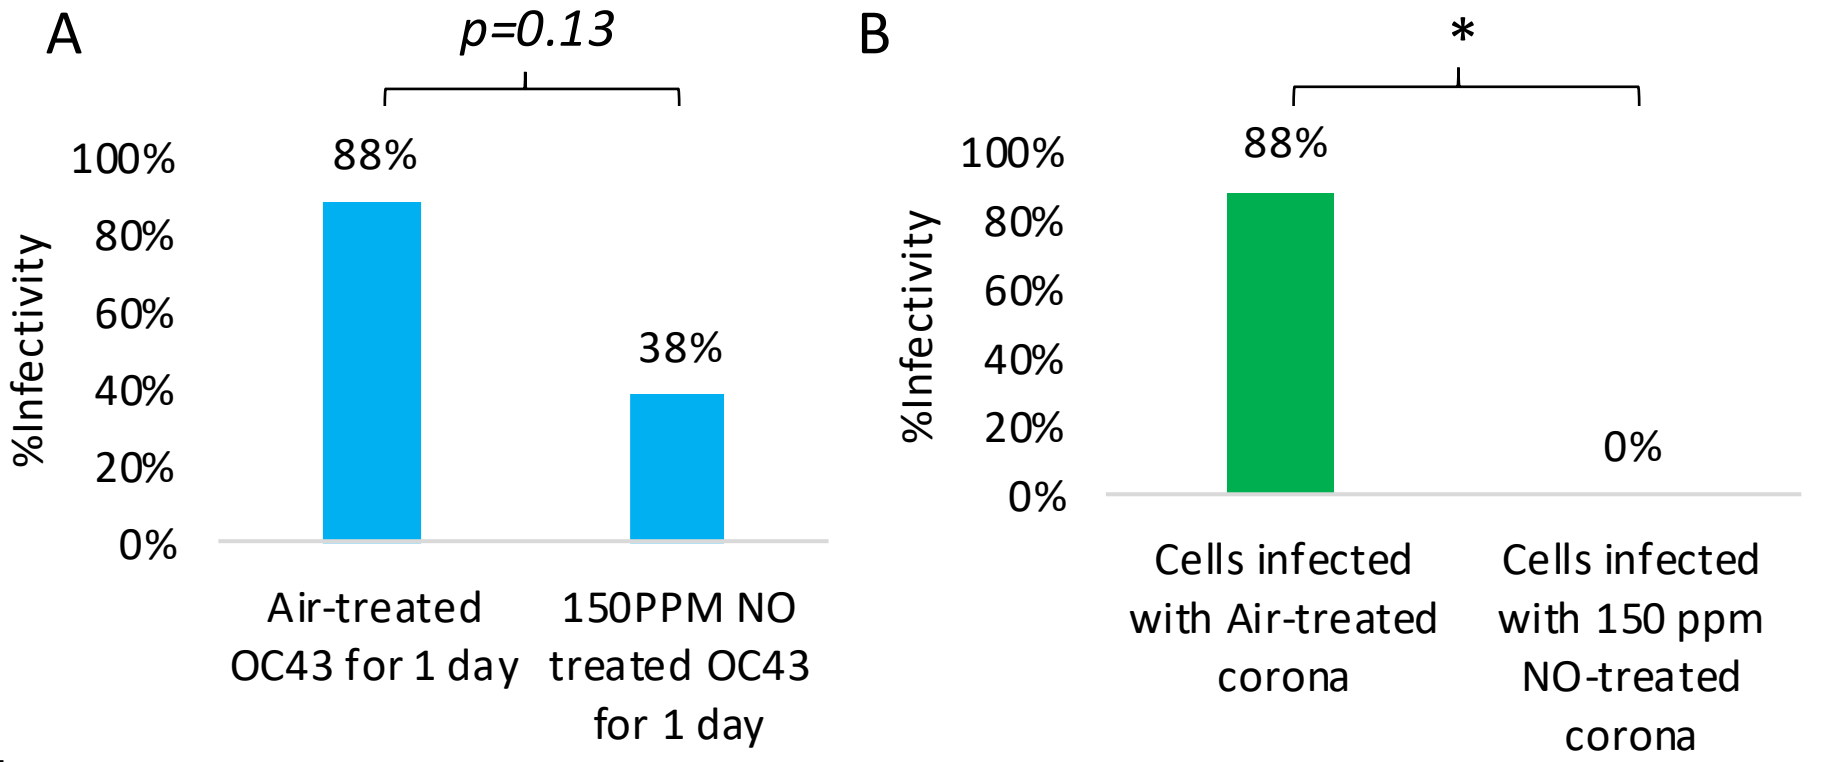
A
%Infectivity
0%
20%
40%
60%
80%
100%
p=0.13
88%
38%
Air-treated OC43 for 1 day
150PPM NO treated OC43 for 1 day
B
%Infectivity
0%
20%
40%
60%
80%
100%
*
88%
0%
Cells infected with Air-treated corona
Cells infected with 150 ppm NO-treated corona

Figure 2: Exposure of OC43 coronavirus infected cells to 150 ppm NO reduces virus infectivity. Infectivity of OC43 human coronavirus extracted from host cells treated with 150 ppm NO or air, 4 cycles of 1-hour exposure to NO and 1-hour in the incubator for A) 1 day of treatment and B) 2 days of treatment. (McNemar Chi square test, n=8 pairs, *$p<0.05$)

Pre-exposure of OC43 human coronavirus to 250 ppm NO reduces viral infectivity

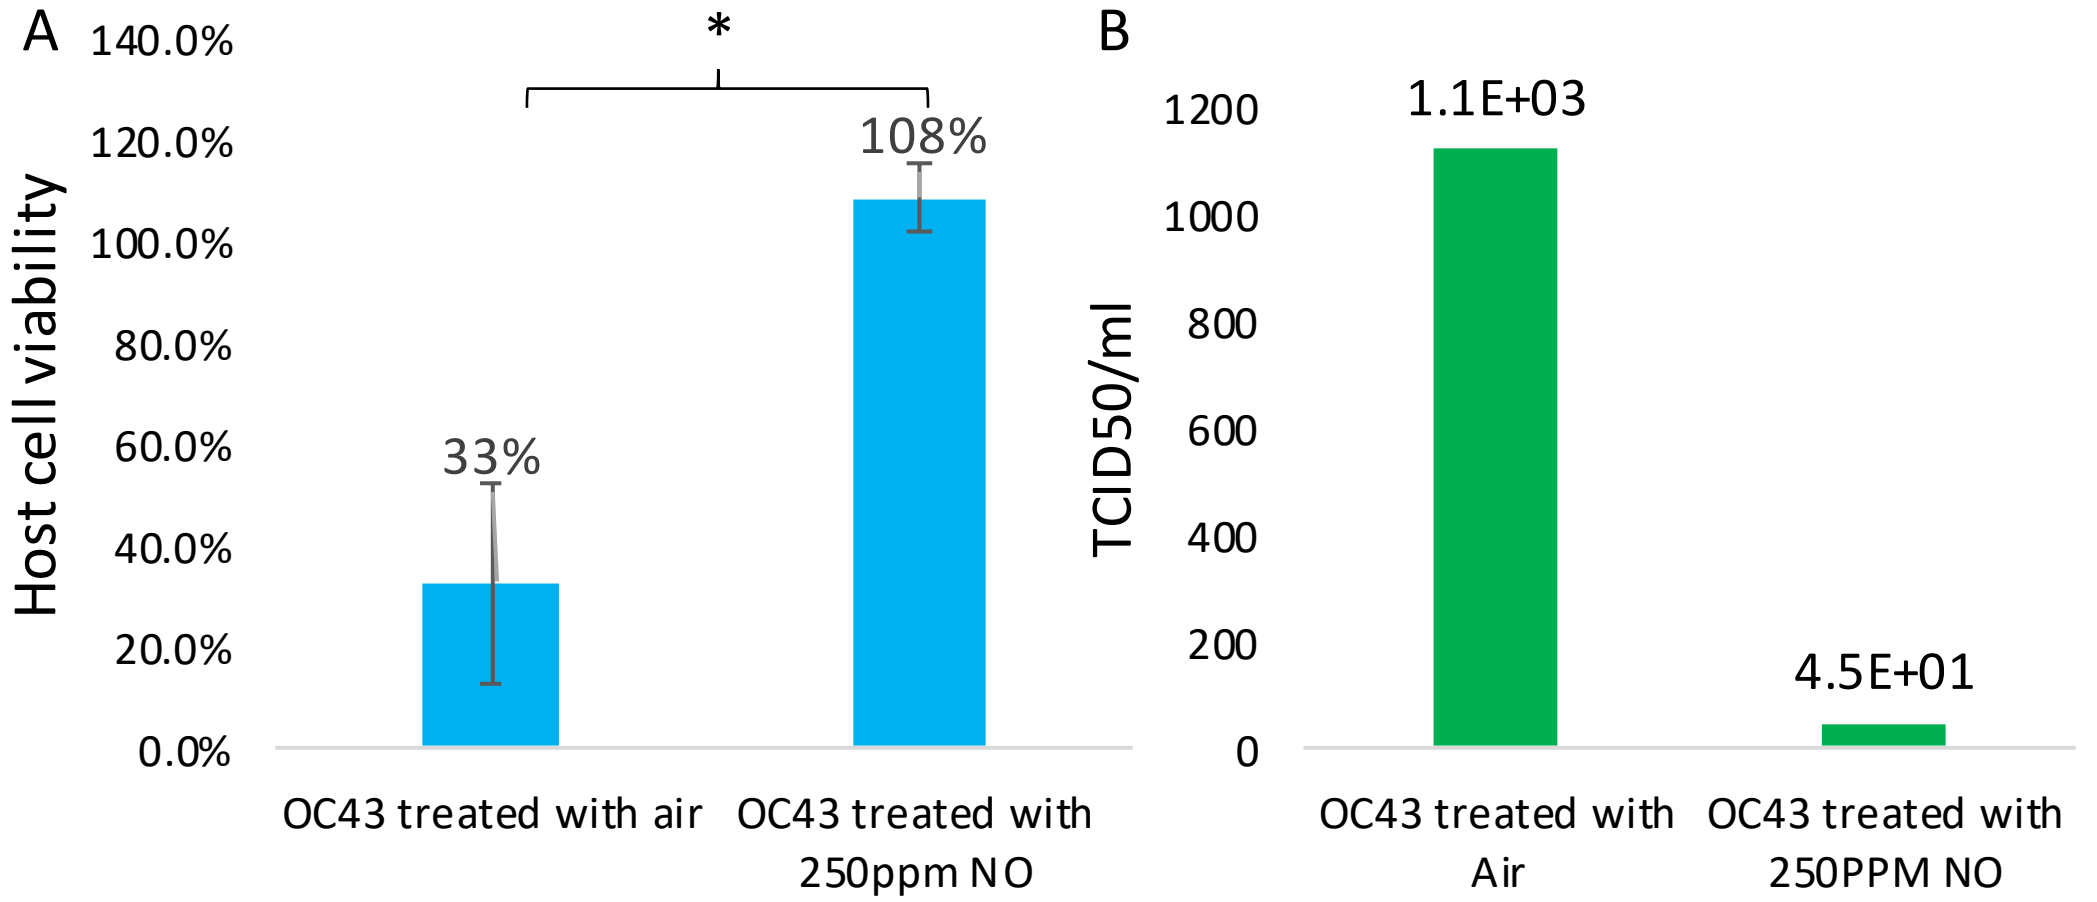
A
Host cell viability
0.0%
20.0%
40.0%
60.0%
80.0%
100.0%
120.0%
140.0%
*
33%
108%
OC43 treated with air
OC43 treated with 250ppm NO
B
TCID50/ml
0
200
400
600
800
1000
1200
1.1E+03
4.5E+01
OC43 treated with Air
OC43 treated with 250PPM NO

Figure 3: A single 2-hour exposure to 250PPM NO prior to infection reduces OC43 infectivity and improves host cell viability. A) Viability of HCT-8 cells 7-days after infection with OC43 human coronavirus pre-treated for 2 hours with 250ppm NO or air (n=3, mean±SD, normalized to non-treated cells, *$p<0.05$, Student's t-test) B) TCID50/ml of HCT-8 infected cells with pre-treated OC43 human coronavirus 6 days post-infection.

Conclusions: These *in vitro* results indicate the potential of inhaled gaseous NO as a novel treatment for human coronavirus infection. According to our data, 150-250ppm gaseous NO shows anti-coronavirus properties against OC43 human coronavirus *in vitro* , when administered either prior to or post infection. The data show that the LungFit™ system may be effective for both prevention and treatment of SARS-CoV-2 infection.